Use of Statistical Design and Automation in Optimizing a Protein Tyrosine Phosphatase-1B Activity Assay

Maureen Beresini[1], Sherry Yeh[2], and Saloumeh Kadkhodayan[1]
BioAnalytical Research and Development[1] and Assay and Automation Technology[2], Genentech, Inc. South San Francisco, CA 94080

ABSTRACT

Protein tyrosine phosphatase-1B is a potential therapeutic target for the treatment of type 2 diabetes. Antagonists of this enzyme may inhibit dephosphorylation of the insulin receptor, and consequently, maintain it in an activated state leading to increased insulin sensitivity. To evaluate the activity of potential antagonists of this enzyme, a fluorescein diphosphate-based activity assay was developed. Performance of this assay was variable; furthermore, both enzyme and antagonist activity varied with buffer conditions. Therefore, the assay was reoptimized using the Sagian™ Automated Assay Optimization system on a Beckman BioMek® 2000. Ten buffer factors were evaluated in an iterative fashion for their effects on assay performance as assessed in terms of total activity, background activity, inhibition by antagonist, and signal-to-noise ratio. Enzyme and substrate concentrations were then optimized in the refined buffer system. Statistical design proved to be a powerful tool for efficient optimization of this assay, and automation greatly facilitated its application.

INTRODUCTION

Protein Tyrosine Phosphatase-1B (PTP-1B)

PTP-1B is a key modulator of insulin sensitivity; it dephosphorylates the insulin receptor, and consequently, returns it to an inactive state. Inhibitors of this enzyme may serve to maintain the insulin receptor in an activated state, and therefore, may have therapeutic potential in the treatment of type 2 diabetes. Efforts were underway to identify peptides which bind to PTP-1B and also to generate small molecule inhibitors using structure based design. Assays which had been developed to support this project include a fluorescein diphosphate (FDP)-based activity assay, a peptide-based activity assay, an insulin receptor kinase receptor activitation (KIRA) assay, and a fluorescence polarization peptide binding assay. In the FDP-based activity assay, we noted atypical variability in both the enzyme activity level and the level of inhibition by antagonists. Furthermore, the assay was very sensitive to buffer conditions. Therefore, we considered this assay a good candidate for reoptimization using the Sagian™ Automated Assay System.

Sagian™ Automated Assay Optimization (AAO)

AAO is an integrated system of hardware and software for optimizing assay conditions. It combines statistical design of experiments based on 2-level designs with the automated pipetting of the Biomek® 2000 (a single-channel liquid-handling workstation). The software employs a step-by-step wizard-like interface to set up the experimental design and establish the protocol parameters for the Biomek® 2000.

Biomek® 2000

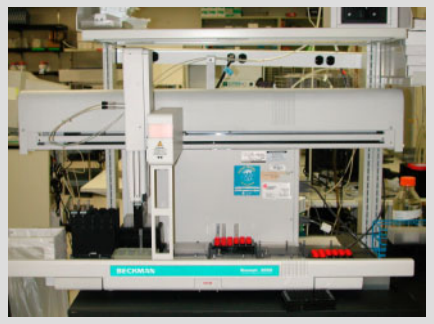

AAO involves 5 main steps:

Design the experiment
•Identify factors and levels
•Select the design resolution
•Specify the protocol

Generate the plate maps

Create the BioWorks methods
•Specify deck layout, pipetting parameters, labware assignments, sequence of addition

Perform the assay
•Run the Biomek method
•Provide manual coordination

Collect and analyze the data

INTRODUCTION (continued)

Statistical Design of Experiments (DOE) in Assay Optimization

DOE facilitates assay optimization by allowing the examination of a large number of variables in an efficient manner. The use of statistics is critical in ensuring appropriate design of the experiments and in interpreting the data.

An integrated strategy for assay optimization follows:

Specify the goal for the optimization
•Identify the important responses

Identify factors of biological relevance to the assay
•Both quantitative and qualitative
•From the literature and/or current practice

Begin with a screening strategy
•Two-level fractional factorial design

Focus progressively on optimization
•Identify main effects
•Determine if there are significant interactions
•Optimize the levels of the important factors

Verify optimized conditions
•Determine kinetic constants
•Evaluate performance

Two-Level Factorial Designs
Designs in which each factor is examined at 2 levels
•Simplify experimentation and analysis
•Require 2^f experiments, where f is the number of factors
•Indicate major trends
•Are blind to curvature
•Can detect interactions
•Represent balanced designs
-Half of the experiments are run at each level of each factor

Fractional Factorial Designs
Designs which use only a fraction of the experiments required for a full factorial design
•Allow to reduce the number of experiments
•Allow to distinguish the important factors from the trivial
-Pareto Principle
•$1/2^p$ denotes the level of fractionation
•Number of experiments = 2^{f-p}
•Ability to detect interactions decreases as the extent of fractionation increases
•Design resolution refers to the ability to distinguish main effects from each other and from multi-factor interactions.
•It is important to balance the ability to resolve interactions with the number of experiments required

Main Effects
•Alterations in the response produced by individual factors
•Effect estimate: the difference between the means of the high level responses and the low level responses for a given factor

Interactions
•Result when the response to the combination of two factors is not the expected sum of their individual effects
•Effect estimate: half of the difference between the effects of one factor at each of the two levels of the other factor
•Interactions generally decrease in magnitude as the number of factors involved increases, so two-factor interactions are the primary concern

Sample Design Matrix: 2^{5-2} (One-Quarter Fraction)

	Settings				
	A	B	C	D ABC	E BC
Expt #	PEG	Glycerol	NaCl	CHAPS	EDTA
3	-	-	-	-	+
6	-	-	+	+	-
8	-	+	-	+	-
2	-	+	+	-	+
7	+	-	-	+	+
5	+	-	+	-	-
1	+	+	-	-	-
4	+	+	+	+	+

of Factors: 5
of Experiments: 8
Resolution: III
Design Generators: D=ABC, E=BC
Two-Factor Aliases: A=DE, B=CE, C=BE, D=AE, E=AB, AB=CD, AC=BD

MATERIALS & METHODS

Key Reagents

Enzyme: PTP-1B, recombinant human, amino acids 1-321, produced in E. coli (Genentech)
Substrate: Fluorescein diphosphate (Molecular Probes)
PTP-1B Inhibitor: Suramin (Calbiochem)

Fluorescein Diphosphate-Based Activity Assay

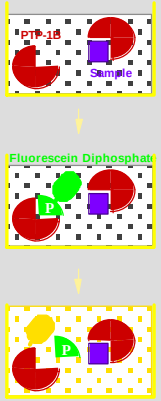
PTP-1B
Sample
Fluorescein Diphosphate
P

10 µL sample in 30 % DMSO
+
40 µL PTP-1B
Incubate at RT for 15 min
50 µL Fluorescein Diphosphate
Mix
Read fluorescence every 30 sec for 10 cycles (ex. 485 nm / em. 535 nm)

The kinetic activity assay uses FDP as substrate. The assay protocol was modified slightly for use within the AAO system. A combination of manual and automated procedures were employed to facilitate the assay runs. The experiments were designed and randomized in AAO; this resulted in the creation of a BioWorks method for operating the Biomek® 2000. Generally, the test buffers were prepared on the Biomek® 2000. Ninety microliters of each of the test buffers were added to the appropriate wells of duplicate, polypropylene, round-bottom plates. One of these plates was used for preincubation of sample and enzyme and the other plate was used for dilution of substrate. 12.5 µL of 60 % DMSO (total or background) or 50 µM suramin in 60 % DMSO (standard) and 12.5 µL of enzyme (total or standard) or of buffer (background) were added to the wells of the first dilution plate, and 10 µL of fluorescein diphosphate were added to the wells of the second plate. If the FDP concentration was constant, the FDP was added to the wells manually with a multi-channel pipet. All other additions to these plates were made on the BioMek® 2000. The contents of each plate were mixed gently by trituration with a multi-channel pipet.

The enzyme and sample (60 % DMSO +/- suramin) were preincubated for a minimum of 15 minutes. To start the reaction, 50 µL of the enzyme-sample solution followed 50 µL of the diluted substrate were manually added to the corresponding wells of a black 96-well assay plate (Costar). The contents of the plates were mixed for 15 seconds on a rotating shaker, and the plates were read kinetically on a Wallac Victor II (excitation 485 nm; emission 535 nm; read every 30 sec for a total of 10 cycles).

The following table summarizes the 5 experimental runs that were used to optimize the assay conditions:

Run #	1	2	3	4	5
# of Factors	10	5	3	3	2
# of Buffer Factors	10	5	3	3	0
# of Levels/Factor	2	2	2	3	5
# of Wells	192	192	192	162	150
# of Replicates	1	2	8	2	3
Design Resolution	IV	Full	Full	Full	Full
Protocol	T, S, B	T, S, B	T,S,B	T,S,B	T,S,B

The first four runs were used to optimize the buffer components, and the fifth run was used to optimize the concentrations of enzyme and substrate. Runs # 4 and 5 were designed in JMP 4.0 and imported into AAO. All runs include total (T), standard (S; 10 µM suramin), and background (B) wells. For runs # 1-4, enzyme and substrate concentrations were 2 nM and 40 µM, respectively. The data were processed in either AAO (runs # 1-3) or JMP 4.0 (runs # 4 & 5). The responses for runs # 1-3 are displayed as Pareto plots which indicate direction and magnitude of main effects and/or two-factor interactions.

RESULTS & DISCUSSION

Assay Optimization Run #1: Buffer Component Screen

Goal: to screen a large number of buffer factors to identify those which are important for optimal activity and inhibition

ID	Factor	-	+	Units
A	BSA	0	0.1	%
B	BGG	0	0.1	%
C	PEG	0	0.1	%
D	Reducing Agent	DTT	ß-ME	
E	NaCl	0	100	mM
F	EDTA	0	5	mM
G	Glycerol	0	5	%
H	CHAPS	0	0.03	%
I	Tween-20	0	0.05	%
J	Base Buffer	Tris pH 7.5	Succ pH 6.0	

Total and Background Pareto

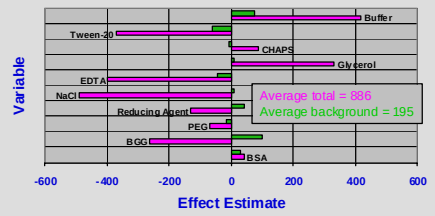
Buffer
Tween-20
CHAPS
Glycerol
EDTA
NaCl
Reducing Agent
PEG
BGG
BSA
Average total = 886
Average background = 195
-600
-400
-200
0
200
400
600
Effect Estimate
Variable

% Inhibition Pareto

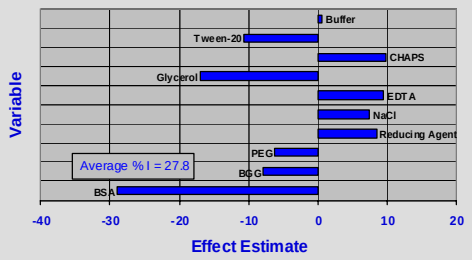
Buffer
Tween-20
CHAPS
Glycerol
EDTA
NaCl
Reducing Agent
PEG
BGG
BSA
Average % I = 27.8
-40
-30
-20
-10
0
10
20
Effect Estimate
Variable

<u>**Buffer Factors to Eliminate from Further Consideration**</u>:
BSA: Minimal effect on signal magnitude; strong negative effect on % inhibition
BGG: Reduced the total signal, increased the background signal, and reduced % inhibition
NaCl, EDTA, Tween-20: Very strong negative effects on total signal

<u>**Buffer Factors for Further Evaluation**</u>:
PEG: Slightly reduced total signal, background signal, and % inhibition; the least negative of the bulking agents studied
Reducing agent: DTT was favorable to ß-ME with respect to total and background signal but not % inhibition
Glycerol: Strong positive effect on total signal; strong negative effect on % inhibition
CHAPS: Positive effects on both total signal and % inhibition
Base buffer: Succinate had a strong positive effect on total signal; little effect of base buffer on % inhibition

Run #2: Buffer Component Optimization

Goal: to more fully characterize the main effects and interactions of the important buffer factors from run #1 in attempt to maximize total signal and percent inhibition

ID	Factor	-	+	Units
A	Base Buffer	Tris pH 7.5	Succ pH 6.0	
B	CHAPS	0	0.03	%
C	Glycerol	0	5	%
D	Reducing Agent	DTT	ß-ME	
E	PEG	0	0.1	%

Total and Background Pareto

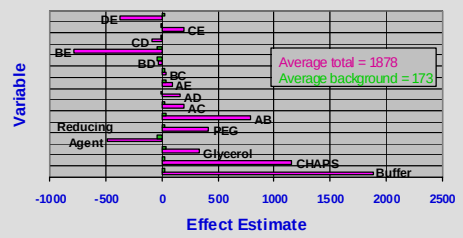
DE
CE
CD
BE
BD
BC
AE
AD
AC
AB
Reducing Agent
PEG
Glycerol
CHAPS
Buffer
Average total = 1878
Average background = 173
-1000
-500
0
500
1000
1500
2000
2500
Effect Estimate
Variable

Succinate, CHAPS, glycerol, DTT, and PEG each have positive effects on assay signal. Nevertheless, interactions between factors need also be considered. There is a strong positive interaction between the buffer and CHAPS (AB); however more importantly, there is a strong negative interaction between CHAPS and PEG (BE).
Succinate and DTT were selected over Tris and ß-ME for further studies.

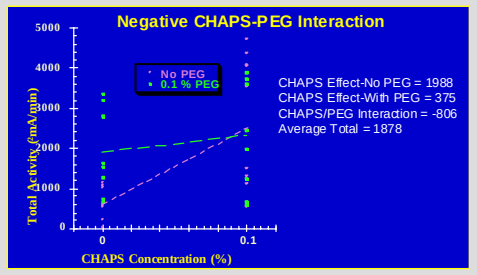
Negative CHAPS-PEG Interaction
No PEG
0.1 % PEG
CHAPS Effect-No PEG = 1988
CHAPS Effect-With PEG = 375
CHAPS/PEG Interaction = -806
Average Total = 1878
Total Activity (mA/min)
5000
4000
3000
2000
1000
0
0
0.1
CHAPS Concentration (%)

Run #3: Continued Buffer Component Optimization

Goal: to further evaluate the effects of CHAPS, glycerol, PEG, and the CHAPS-PEG interaction on total signal, % inhibition, and precision (n=8)

ID	Factor	-	+	Units
A	CHAPS	0	0.03	%
B	Glycerol	0	5	%
C	PEG	0	0.1	%

Base buffer contains 50 mM succinate pH 6.0 and 2 mM DTT

% Inhibition Pareto

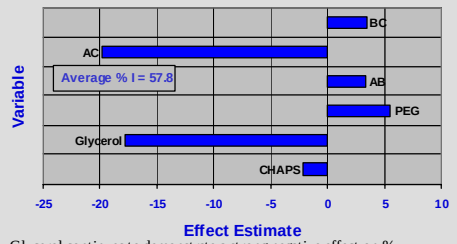
BC
AC
AB
PEG
Glycerol
CHAPS
Average % I = 57.8
-25
-20
-15
-10
-5
0
5
10
Effect Estimate
Variable

Glycerol continues to demonstrate a strong negative effect on % inhibition. The CHAPS-PEG (AC) interaction is negative for % inhibition as well for total signal.

SignalNoiseRatio Pareto

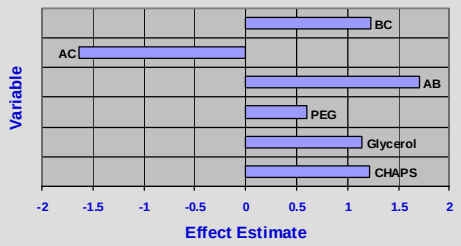
BC
AC
AB
PEG
Glycerol
CHAPS
-2
-1.5
-1
-0.5
0
0.5
1
1.5
2
Effect Estimate
Variable

CHAPS, glycerol, and PEG each have positive effects on signal-to-noise ratio (S/N), and the interactions of glycerol with CHAPS (AB) or PEG (BC) are also positive, so glycerol has the expected positive effect on precision. Nevertheless, the negative interaction between CHAPS and PEG (AC) also extends to the signal-to-noise ratio.

Run #4: 3-Level Buffer Factor Optimization

Goal: to use a 3-level design to determine whether intermediate concentrations of CHAPS, PEG, and/or glycerol may lead to more optimal responses

Factor	-	0	+
CHAPS	0	0.01 %	0.03 %
PEG	0	0.01 %	0.1 %
Glycerol	0	1 %	5 %

CHAPS	PEG	Glycerol	Total	Standard	Background	T/B	S/N
0.03	0	0	3311	1231	594	5.8	3.6
0.01	0	0	3951	1133	241	19.1	8.0
0	0.1	1	3047	1072	245	12.9	8.2
0.01	0.01	0	3824	927	429*	11.5	10.7
0.01	0.01	1	3881	1338	278*	24.5	7.1

*Background signal values were very discrepant between the two plates; background was consistently and considerably higher on the second of two plates.

The conditions resulting in the best set of responses in the previous run were 0.03 % CHAPS without PEG or glycerol. The results from these conditions for this run are shown in red. The results of the four other conditions yielding the best responses for this run are also included in the above table. All of these four conditions contain at least one of the factors at its intermediate level. The new set of conditions yielding the best set of responses is shown in green. It corresponds to 0.01 % CHAPS and demonstrates improvement across the board as compared to 0.03 % CHAPS.

Run #5: Enzyme and Substrate Concentration Optimization

Goal: to use a 5-level design to optimize enzyme and substrate concentrations to yield the best combination of total activity and percent inhibition

Factor	1	2	3	4	5
Enzyme Concentration (nM)	0	0.5	1	2	4
Substrate Concentration (µM)	10	20	40	60	80

Optimized Buffer: 50 mM succinate, 0.01 % CHAPS, 2 mM DTT, pH 6

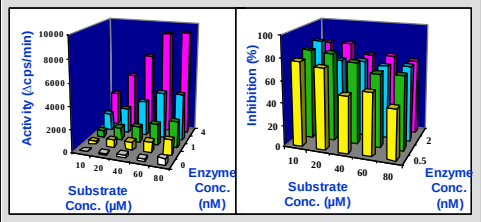
Activity (Δcps/min)
Substrate Conc. (µM)
Enzyme Conc. (nM)
Inhibition (%)
Substrate Conc. (µM)
Enzyme Conc. (nM)

Total signal increases with enzyme concentration as expected. It also increases with substrate concentration up to about 60 µM, and then it levels off. Substrate concentration has a negative effect on % inhibition, and enzyme concentration has a positive effect. That is, inhibition increases with enzyme concentration and decreases with substrate concentration. Choosing a high enzyme concentration should be good for both total activity and % inhibition, so 2 and 4 nM enzyme concentrations were further evaluated with respect to Km determination and suramin inhibition curves.

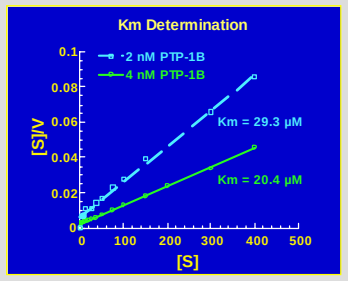
Km Determination
2 nM PTP-1B
4 nM PTP-1B
Km = 29.3 µM
Km = 20.4 µM
[S]/V
[S]
0
100
200
300
400
500

Several sets of suramin titration curves were run at 2 and 4 nM enzyme in combination with 20 to 60 µM substrate (1 - 2x the Km). The IC_{50} values varied by less than 2-fold across the set of conditions examined. The curves for the two best sets of conditions are shown below. Both of these sets of conditions were carried into a final study to evaluate assay performance.

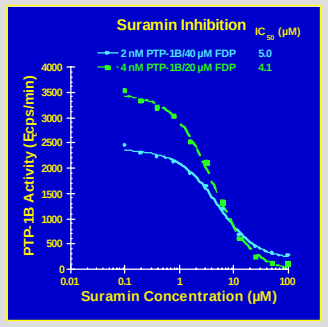
Suramin Inhibition
IC50 (µM)
2 nM PTP-1B/40 µM FDP 5.0
4 nM PTP-1B/20 µM FDP 4.1
PTP-1B Activity (Δcps/min)
Suramin Concentration (µM)
0.01
0.1
1
10
100

Performance Evaluation

Enzyme Conc. (nM)	Substrate Conc. (µM)	Total	Standard	Back-ground	% Inhibition	T/B	S/N	Z'-Factor
2	40	1807	798	266	65.5	6.8	7.6	0.51
4	20	3047	786	92	76.5	33.1	9.7	0.67

*n = 12 in each of 3 assay runs

4 nM enzyme with 20 µM substrate is more favorable across the board: higher total activity, lower background, greater % inhibition, greater T/B, greater S/N, and greater Z'-factor.

<u>**Optimized Conditions**</u>:
50 mM succinate, 0.01 % CHAPS, 2 mM DTT, pH 6
4 nM PTP-1B
20 µM fluorescein diphosphate

SUMMARY / CONCLUSIONS

•Statistical design is a powerful tool for assay optimization
•Automation facilitates its application
•Together statistical design and automation facilitate the coverage of a greater amount of experimental space than can be covered by traditional methods
•The AAO system is best suited for 2-level designs aimed at identifying the important factors and interactions
•Additional software packages are required for higher level designs and their analysis
•The AAO system was successfully used for optimizing the PTP-1B activity assay through an iterative process of screening and optimization followed by verification

ACKNOWLEDGEMENTS:

Randy Yen, Wai Lee Wong, Kurt Deshayes, Mark Renz, Barbara Moffat, Mark Ultsch, Ellen Gilkerson, Laurie Myers, and Duane Bloedow

REFERENCES:

1. Elchebly M, Payette P, Michaliszyn E, Cromlish W, Collins S, Loy AL, Normandin D, Cheng A, Himms-Hagen J, Chan C-C, Ramachandran C, Gresser MJ, Tremblay ML, and Kennedy BP. Increased Insulin Sensitivity and Obesity Resistance in Mice Lacking the Protein Tyrosine Phosphatase-1B Gene. Science 283:1544-1548 (1999).
2. Morgan, Ed. Chemometrics: Experimental Design. John Wiley & Sons Ltd., West Sussex, England, 1995.
3. Haaland, Perry D. Experimental Design in Biotechnology. Marcel Dekker, Inc., New York, 1989.
4. Sherrill T, Snider J, Terpstra N, Vanderpool C, and Schmidt W. Sagian™ Automated Assay Optimization - A New Solution for Accelerating Assay Development. Technical Information: T-1862A. Beckman Coulter, 1999.
5. Taylor PB, Stewart FP, Dunnington DJ, Quinn ST, Schulz CK, Vaidya KS, Kurali E, Lane TR, Xiong WC, Sherrill TP, Snider JS, Terpstra ND, and Hertzberg RP. Automated Assay Optimization with Integrated Statistics and Smart Robotics. Journal of Biomolecular Screening 5(4):213-225 (2000).